Improved Accuracy of Pathology Results Helps with Kidney Utilization: Comparison of Complex Kidney Biopsy Pathology Results

LifeGift
Donation Optimization
CONSORTIUM

W. Ambroise MSHA, MBA, RRT, RCP, CPTC[1], N. Powley, K. Myer[1], MSHA, Y. Becker[1], MD, V. Lopez-Segura[1], S. Rombach[2], A. Meinders[3], A. Waterman[3], D. Axelrod[4],
LifeGIft Organ Donation Center Houston, TX, Specialist Direct, San Diego, CA, Houston Methodist Hospital Houston, TX, University of Iowa, Iowa City, IA

INTRODUCTION

- The role of kidney transplant procurement biopsies is controversial; however biopsy results are used by transplant centers to inform allograft acceptance for transplantation. Centers and OPOs rely on kidney biopsy interpretation by pathologists on-call at local donor hospitals. Very few pathologists have advanced training in kidney interpretation[1]. This may contribute to an overestimate of percentage of glomerulosclerosis and thereby a higher non-use rate despite a paucity of data to support this approach[2]. Unfortunately access to 24/7 fellowship-trained renal pathologists to interpret the biopsies is limited. We present a comparison of the times zero donor kidney biopsies were read by the on-call pathologist vs. telepathology readings that were completed by a fellowship trained pathologist.

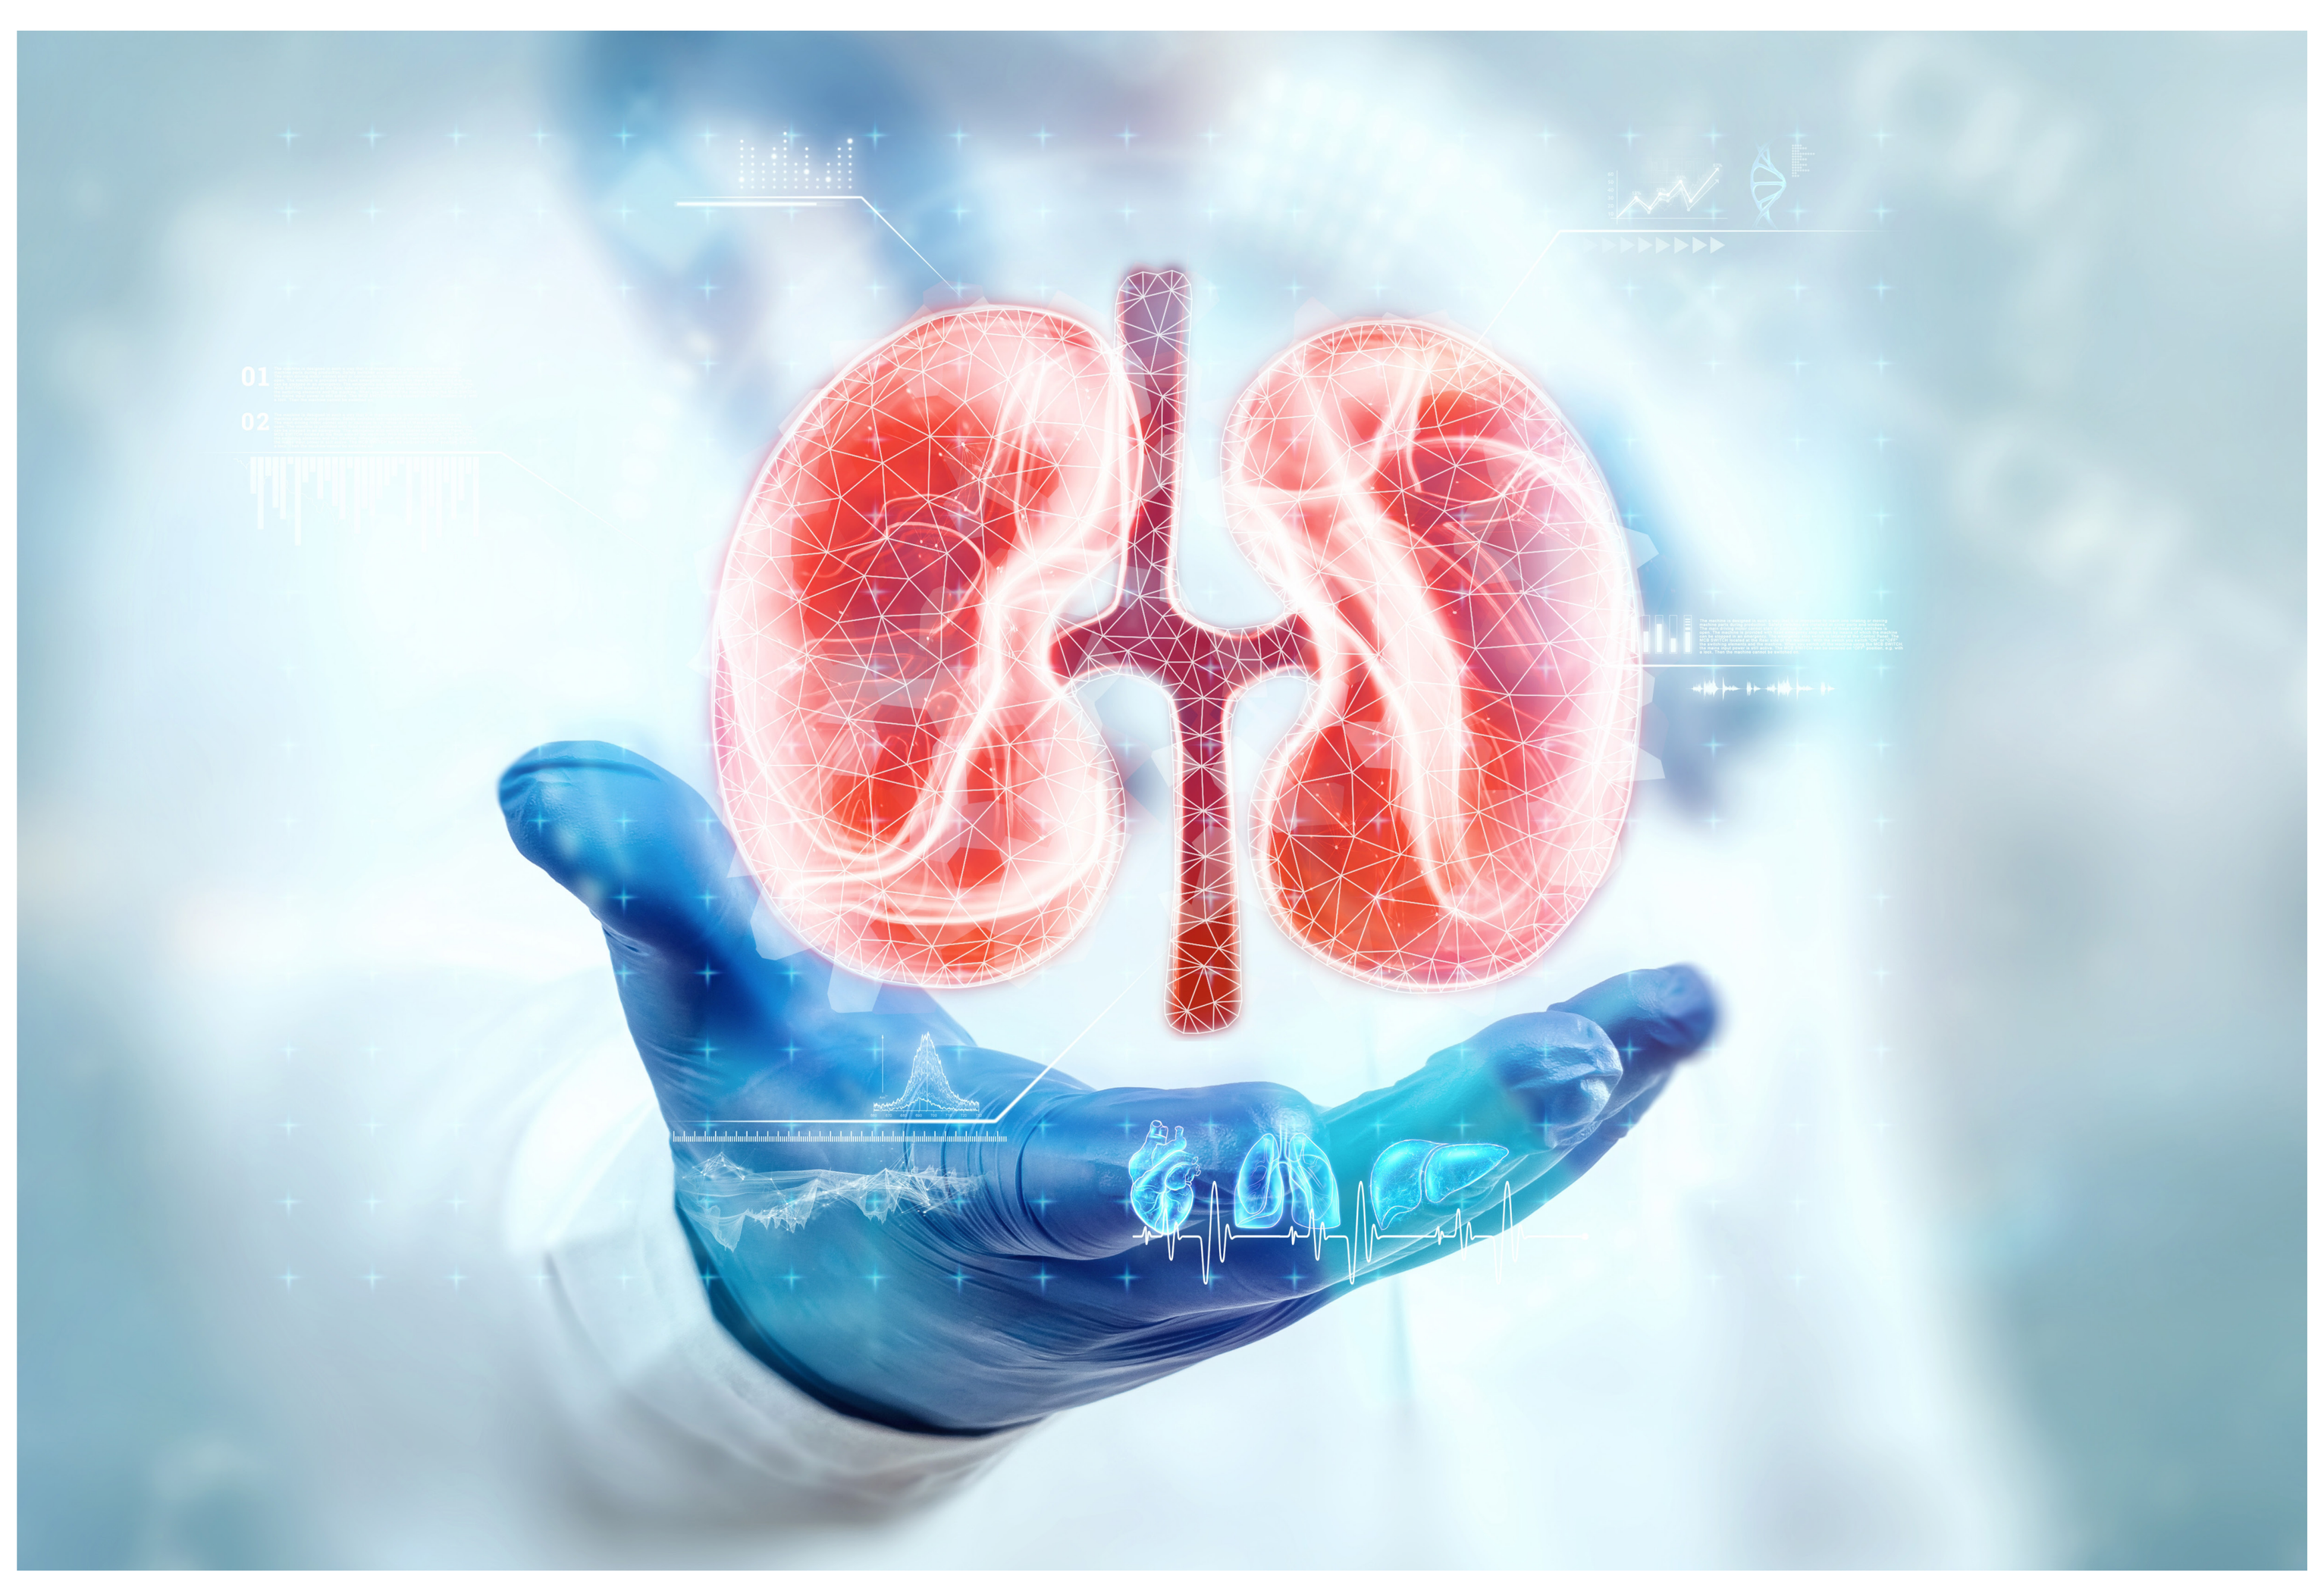

This work was supported wholly or in part by HRSA contract 75R60220C00011. The content is the responsibility of the authors alone and does not necessarily reflect the views or policies of the Department of HHS, nor does mention of trade names, commercial products, or organizations imply endorsement by the U.S. Government.

There are no conflicts of interest, and we have nothing to disclose.

METHODS

- We compared onsite interpretations of procurement kidney biopsies in real time vs. readings performed via telepathology.
- Complete pathology readings included the number of glomeruli seen, percent glomerulosclerosis number, severity of fibrosis and vascular disease.
- The initial and final kidney transplant outcome was also reviewed.

Figure A

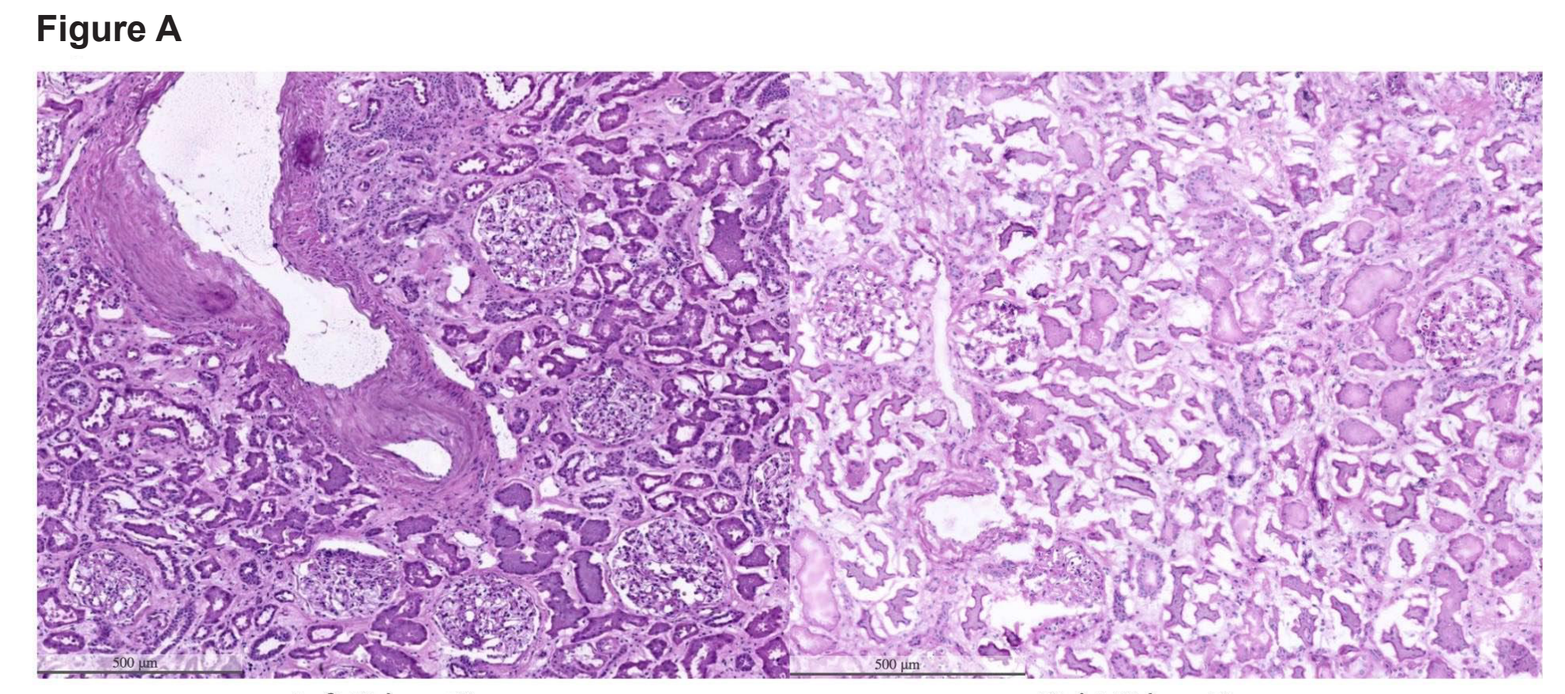

Left Kidney Biopsy Right Kidney Biopsy

	Local Pathologist		Standardized Pathologist	
	Left Kidney	Right Kidney	Left Kidney	Right Kidney
Glomeruli Seen, n	> 100	> 100	146	141
Glomeruli Sclerosis, n	3	2	4	4
Sclerosis, %	3%	2%	3%	3%
Interstitial Fibrosis	Absent	Absent	Mild	Mild
Arteriosclerosis	No	No	Mild	Mild
Vascular Changes	Absent	Minimal	None	None
Other	None	None	Simple cysts	Simple cysts
Outcome			Transplanted	Transplanted

Figure B

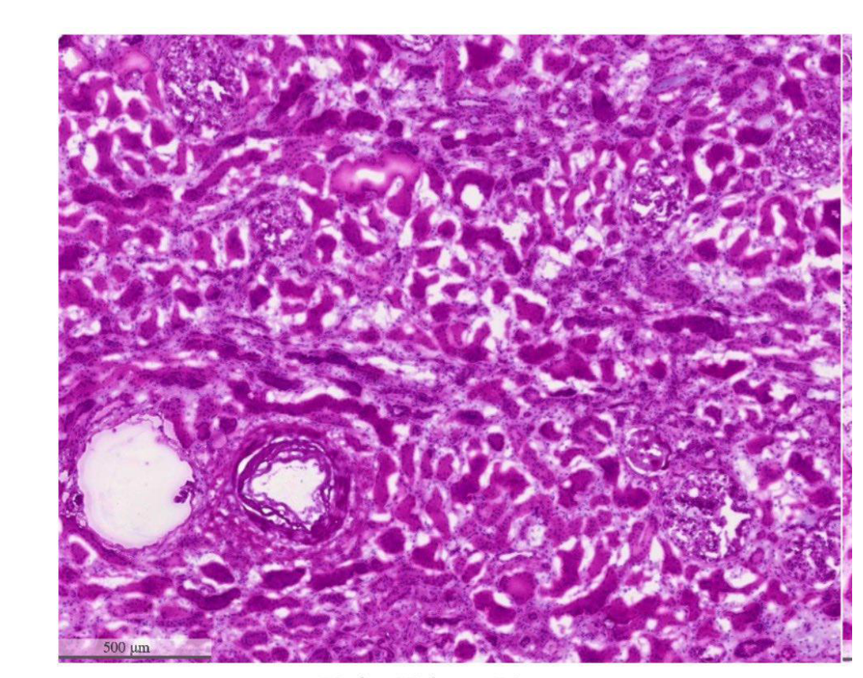

Left Kidney Biopsy Right Kidney Biopsy

	Local Pathologist		Standardized Pathologist	
	Left Kidney	Right Kidney	Left Kidney	Right Kidney
Glomeruli Seen, n	--	63	--	91
Glomeruli Sclerosis, n	--	29	--	13
Sclerosis, %	--	46%	--	14%
Interstitial Fibrosis	--	Severe	--	6-25%
Arteriosclerosis	--	26-50%	--	26-50%
Vascular Changes	--	Severe	--	Moderate
Other	--	None	--	None

Figure C

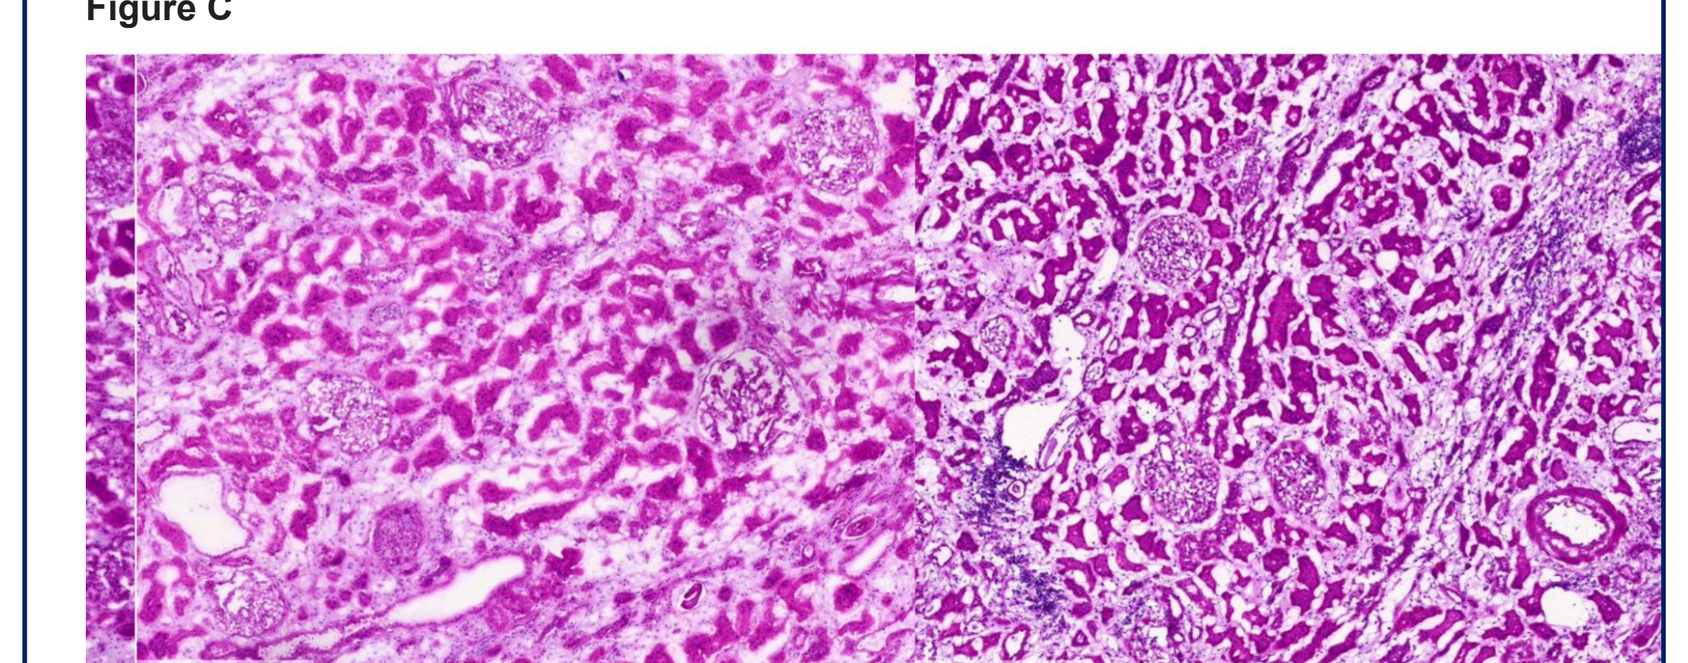

Left Kidney Biopsy Right Kidney Biopsy

	Local Pathologist		Standardized Pathologist	
	Left Kidney	Right Kidney	Left Kidney	Right Kidney
Glomeruli Seen, n	144	127	116	168
Glomeruli Sclerosis, n	13	15	6	20
Sclerosis, %	9%	12%	5%	12%
Interstitial Fibrosis	Severe	Severe	25-50%	6-25%
Arteriosclerosis	>50%	11-25%	26-50%	10-25%
Vascular Changes	Severe	Mild	Moderate	Mild
[illegible]	Chronic [illegible]	Chronic [illegible]	Patchy mononuclear cells [illegible]	Patchy mononuclear cells [illegible]

DISCUSSION

- Our results show that experienced pathologists often have a different interpretation of a donor kidney biopsy than pathologists with limited experience in reading kidney biopsies. Standardized pathology ensures a consistent evaluation and reading. This consistency is essential to provide accurate and reproducible results to transplant centers. The expertise of pathologists accustomed to reading kidney biopsies decreases misinterpretation and increases reliability and trust in the OPO by the transplant centers.

CONCLUSIONS

- The reported severity of sclerosis and fibrosis was higher for local pathologists in contrast to the telepathology interpretation.
- Improved access to photomicrographs through telepathology increases the opportunity to access a single service for consistent interpretation of donor biopsies.
- Engaging a qualified renal pathologist to interpret biopsy slides enhances the accuracy of reading and subsequent acceptance of organs for transplant by kidney transplant centers.

REFERENCES

1. Jhaveri KD, Schmidt IM et al. A Qualitative Evaluation of Advanced Training Programs in Glomerular Diseases: Results from a Program Director's Survey, Kidney Int Rep 2023 Nov; 8(11): 2183-2186.
2. Wong CJ, Wetmore JB et al. Impact of Donor Kidney BIopsy on Kidney Yield and PostTransplant Outcomes, AJT 2023 Mar 23(3):387-392.

LifeGift.org | LifeGift

© LifeGift 2024